CARIPRAZINE & CLOZAPINE
A promising combination for treatment-resistant schizophrenia

Sofia Pappa[1,2], Zsófia B. Dombi[3], Ágota Barabássy[3], Réka Csehi[3], Stephan Hjorth[4]

[1] Faculty of Medicine, Imperial College London, London, UK
[2] West London NHS Trust, London, UK
[3] Global Medical Division, Gedeon Richter Plc., Budapest, Hungary
[4] Pharmacilitator AB, Vallda, Sweden

OBJECTIVE

To evaluate the **real-world effectiveness** of the **cariprazine-clozapine** combination for patients with TRS and explore its potential and benefits from a **pharmacological** point of view.

CONCLUSIONS

By targeting different receptors, cariprazine and clozapine appear to act synergistically.

Emerging real-world evidence also supports this notion with remarkable improvements in TRS patients.

Presented at the 23rd WPA World Congress of Psychiatry, 28 September – 1 October, 2023, Vienna

INTRODUCTION

- Treatment with clozapine is the gold standard when it comes to treatment-resistant schizophrenia (TRS). Nonetheless, partial response characterized by residual negative and cognitive symptoms is common.
- Cariprazine is a 3rd generation antipsychotic medication with a unique receptor profile and proven efficacy in persistent negative symptoms.
- The combination of these two antipsychotics might provide a viable solution for patients with insufficient response to clozapine.

METHODS

- We performed a systematic literature search on PubMed, looking for English language articles published between January 2017 - February 2023 with the following search terms: cariprazine AND clozapine.
- Titles and abstracts of identified articles were screened to determine eligibility; studies and case reports involving cariprazine-clozapine combination treatment in patients with schizophrenia were included.

RESULTS

- Out of the 39 articles, 6 met the inclusion criteria: 1 pilot study and 5 case reports describing 9 and 11 patients, respectively.
- From pharmacological point of view, cariprazine and clozapine have a complementary receptor profile (Figure 1) (1-7).
- Except two patients (10%), all cases had improvement with the add-on cariprazine therapy in positive, negative, and cognitive symptoms (Table 1). Similar results were acquired in the pilot study with significant reduction in the total and the sub-scores of the Positive and Negative Symptom Scale (Table 2).

Figure 1. The receptor profiles of CAR & CLOZ

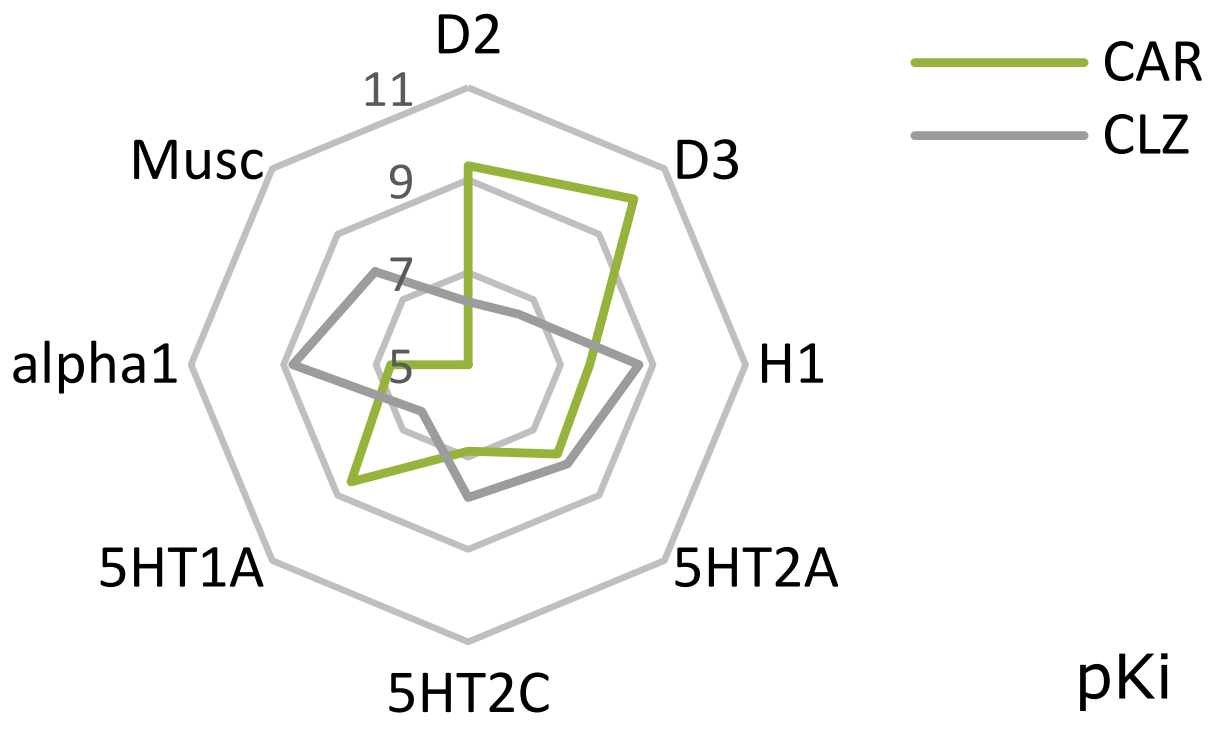

D2
D3
H1
5HT2A
5HT2C
5HT1A
alpha1
Musc
11
9
7
5
CAR
CLZ
pKi

Table 1. Cariprazine-clozapine combinations in case reports

REF.	SEX	AGE	DIAGNOSIS	SYMPTOMS	COMBINATION CAR	COMBINATION CLOZ	OUTCOME
1	M	45	Schizoaffective disorder	Residual negative, cognitive	6.0 mg/d	275 mg/d	Near-complete remission of his persistent negative and psychosocial issues
2	F	29	Schizophrenia	Symptomatic + weight gain	3.0 mg/d	400 mg/d	Symptom improvement, weight loss, found a part-time job
	M	35	Schizophrenia	Symptomatic	3.0 mg/d	300 mg/d	Symptom improvement, found a job
3	F	34	Paranoid schizophrenia	Residual negative	3.0 mg/d	275 mg/d	Improvement in function, reduction in residual positive and negative symptoms
	F	60	Paranoid schizophrenia, depression	Cognitive and negative	1.5 mg/d	250 mg/d	Improvement in negative symptoms
	M	23	Paranoid schizophrenia	Persistent negative	1.5 mg/d	325 mg/d	Improvement in function and persistent negative symptoms
	M	51	Schizoaffective disorder	Persistent negative	1.5 mg/d	600 mg/d	Improvement in function and persistent negative symptoms
	M	28	Paranoid schizophrenia	Persistent negative	1.5 mg/d	700 mg/d	Moderate improvement in negative symptoms
4	M	29	Schizophrenia	Hallucinations and negative	3.0 mg/d	800 mg/d	Disappearance of hallucinations, major improvement in negative symptoms
5	F	41	Treatment-resistant schizophrenia	Recurrent psychotic episodes	6.0 mg/d	350 mg/d	Pisa syndrome, CAR discontinued
	M	63	Treatment-resistant schizophrenia	Obsessive thoughts, cognitive	3.0 mg/d	unknown	Pisa syndrome, CAR discontinued

Table 2. Cariprazine-clozapine combinations in a pilot study

REF.	SEX	AGE	DIAGNOSIS	SYMPTOMS	COMBINATION CAR	COMBINATION CLOZ	OUTCOME PANSS Total - BASELINE	OUTCOME PANSS Total reduction in 3 months
6	M	41	Schizophrenia	Negative + side effects	6.0 mg/d	275 mg/d	24	71%
	M	45	Schizophrenia	Negative + side effects	4.5 mg/d	300 mg/d	39	44%
	M	47	Schizophrenia	Negative + TRS	4.5 mg/d	650 mg/d	102	9%
	F	26	Schizoaffective disorder	Negative + TRS	3.0 mg/d	200 mg/d	62	47%
	F	34	Schizoaffective disorder	Negative + metabolic	1.5 mg/d	200 mg/d	12	67%
	F	33	Schizophrenia + Personality dis.	TRS	6.0 mg/d	200 mg/d	61	79%
	F	31	Schizophrenia + Personality dis.	TRS	6.0 mg/d	200 mg/d	57	60%
	F	34	Schizoaffective + Personality dis.	TRS	1.5 mg/d	500 mg/d	64	Stopped in 6 weeks due to poor response
	M	37	Schizophrenia	Negative + TRS	1.5 mg/d	350 mg/d	92	Stopped in 4 weeks due to restlessness

References: 1. Bogren et al. Front Psychiatry. 2022 May 11;13:887547, 2. Berardis et al. Clin Psychopharmacol Neurosci. 2021 Feb 28;19(1):174-178, 3. Oloyede et al. Ther Adv Psychopharmacol. 2022 Jan 28;12:20451253211066642, 4. Viegas et al. Neuropsychiatr Dis Treat. 2022 Jun 9;18:1145-1149, 5. Weise et al. Clin Psychopharmacol Neurosci. 2022 May 31;20(2):398-401, 6. Pappa et al. Ther Adv Psychopharmacol. 2022 Nov 21;12:20451253221132087, 7. Taylor et al. 2022, International Journal of Psychiatry in Clinical Practice